INT131, a Non-TZD Selective PPARγ Modulator (SPPARM), Does Not Cause Toxicities Typical of TZD Full PPARγ Agonists Following 6-Month High Dose Treatment of Rats or Monkeys

InteKrin THERAPEUTICS

Alex DePaoli and Linda S Higgins

InteKrin Therapeutics, Los Altos, CA

BACKGROUND AND AIMS

PPARγ is a validated target for treatment of insulin resistance, but the available thiazolidinedione (TZD) full agonists have significant side effects. INT131 (formerly T0903131, T131, and AMG 131) is a potent non-TZD selective PPARγ modulator (SPPARM) being developed for treatment of type 2 diabetes mellitus (T2DM). INT131 has demonstrated consistent unique properties in functional assays for receptor activation compared with multiple PPARγ full agonists. Studies with INT131 in rodent models of T2DM have demonstrated similar insulin sensitizing and glucose lowering activity with significantly less fluid retention, weight gain and cardiac hypertrophy than seen with currently available TZDs. A four week clinical study with INT131 in subjects with T2DM has similarly demonstrated significant dose responsive insulin sensitizing and glucose lowering actions, and suggested this efficacy can be separated from typical TZD side effects such as weight gain and fluid retention. Preclinical results for numerous PPAR ligands in rodent and monkey have shown time and dose dependent fluid retention, edema, cardiac hypertrophy, fatty replacement of bone marrow, and in some cases cardiac death. In order to assess the safety profile of INT131, six month safety studies were conducted in cynomolgus monkeys and in rats.

INTRODUCTION

PPARγ is a ligand-activated transcription factor that plays a critical role in the regulation of glucose and lipid homeostasis. PPARγ activation is associated with significant improvement in insulin resistance, hyperglycemia, endothelial function, nonalcoholic steatohepatitis (NASH), polycystic ovarin syndrome (PCOS), and markers of inflammation. INT131 is a novel, selective PPARγ modulator which is not structurally related to the TZD class of PPARγ full agonists and has distinctive binding properties in the nuclear receptor binding pocket (Figure 1). This unique ligand interaction leads to a distinct geometry of INT131 bound PPARγ, which is responsible for association and dissociation of cofactors and corepressors and hence of the transcriptional profile of the activated protein.

Figure 1: Ribbon diagram of INT131 and rosiglitazone in the PPARγ binding pocket demonstrating distinct binding properties, particularly with respect to the activation helix (in red).

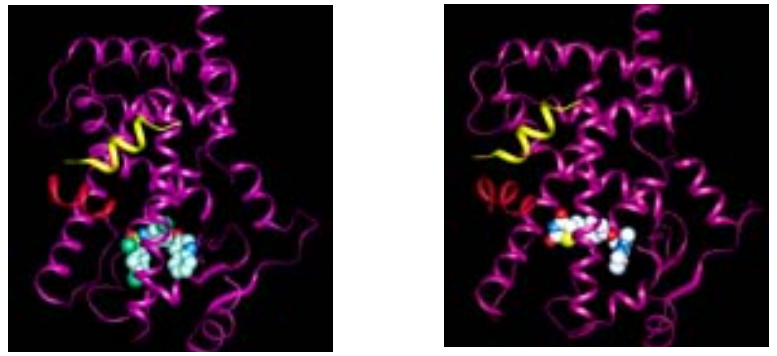
INT131
Rosiglitazone

Full PPARγ activation with the TZDs is associated with a number of undesirable effects that appear to be linked to their beneficial effects in a dose responsive manner. These effects include fluid retention and congestive heart failure (CHF), adipogenic weight gain, and bone resorption associated with fractures. Based on the detailed understanding of PPARγ receptor activation, the opportunity exists to selectively modulate specific activities of the receptor. A Selective PPARγ Modulator (SPPARM) may allow separation of the insulin sensitizing actions of PPARγ activation from the unwanted activities (Figure 2).

Figure 2: Conceptual approaches to selective PPARγ modulation

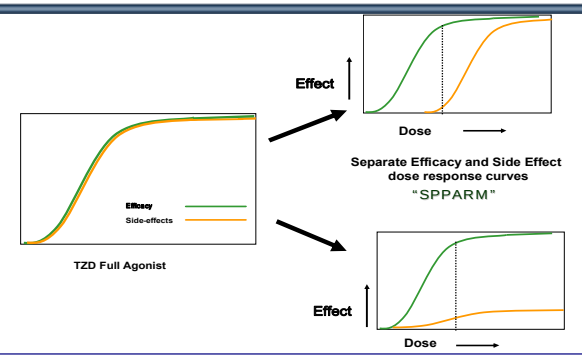
Efficacy
Side-effects
TZD Full Agonist
Effect
Dose
Separate Efficacy and Side Effect dose response curves
"SPPARM"
Effect
Dose

INT131 was specifically designed to retain the anti-diabetic efficacy of PPARγ activation without the unwanted side effects. INT131 has consistently shown SPPARM proof-of-concept from early pharmacology experiments through Phase 2a clinical studies that clearly differentiate it from TZDs and other PPAR full agonists. As shown in Figure 3, INT131 lacks the adipogenic properties of the full agonist TZDs on both rodent and human pre-adipocytes [1]. In Phase 1 and Phase 2a clinical trials, INT131 has demonstrated the ability to safely and robustly stimulate adiponectin and significantly lower FPG [2]

Figure 3: TZDs, but not INT131, are potent inducers of adipogenesis

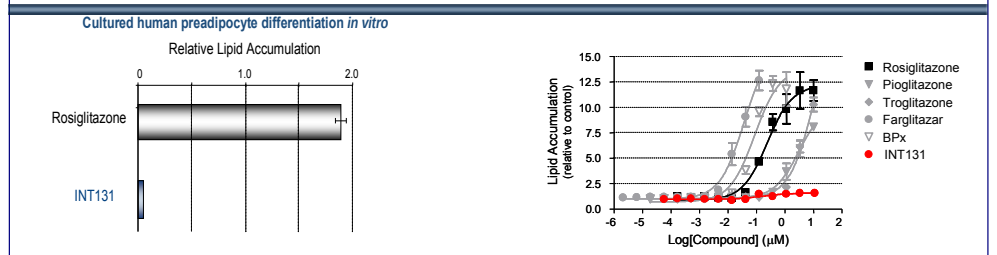
Cultured human preadipocyte differentiation in vitro
Relative Lipid Accumulation
0
1.0
2.0
Rosiglitazone
INT131
Lipid Accumulation (relative to control)
Log[Compound] (μM)
Rosiglitazone
Pioglitazone
Troglitazone
Farglitazar
BPx
INT131

A: Relative lipid accumulation in cultured human pre-adipocytes. Human pre-adipocytes were cultured in the presence of 3 μM rosiglitazone or 3 μM INT131, and assayed after 10 days for differentiation as measured by lipid accumulation with Nile Red staining.

B: Lipid accumulation in murine pre-adipocytes. Cultures were exposed to a range of concentrations of the indicated PPAR ligands. The maximal lipid accumulation stimulated by INT131 was about 10% that promoted by full agonists rosiglitazone, pioglitazone, troglitazone, farglitazar, and BPx.

PPARγ AGONIST PRECLINICAL DATA PREDICT CLINICAL EXPERIENCE

A primary toxicity of concern in both nonclinical safety studies and clinical studies of TZDs is cardiovascular. The cardiac/hematological findings include fluid retention, plasma volume expansion and CHF attributed to the increased preload. In long term rat and mouse studies (e.g., the 2-year carcinogenicity bioassays) the cause of death at the high doses were attributed to cardiac effects. These cardiovascular effects are consistent findings in all nonclinical species tested and occurred at low multiples of clinical exposure. Fluid retention and CHF appear to be a class effects of the TZDs in the clinic, as predicted by the nonclinical studies [3, 4].

TZDs	Rat: Toxicity	Rat: Exposure Ratio (rat:human)	Monkey: Toxicity	Monkey: Exposure Ratio (monkey:human)
Actos ® pioglitazone	**Safety margin** • cardiac hypertrophy • decreased Hct, Hb, RBC • thoracic cavity fluid accumulation • increased lung weight • increased body weight • increased subcutaneous adipose • decreased bone in sternum • **cardiac death**	**4.6 (male)**	**Safety margin** • cardiac enlargement (3 month) • increased food consumption • increased body weight	**4.9**
Avandia ® rosiglitazone	**Safety margin** • cardiac hypertrophy • decreased Hb, RBC, platelet count • thoracic cavity fluid accumulation • atrial thrombi • lung congestion • increased food consumption • increased body weight • **cardiac death**	**5.4 (male)**	Not done	-

* Exposure ratio calculated using approved high dose: 45 mg Actos, 8 mg Avandia.

MATERIALS AND METHODS

Cynomolgus monkeys were treated with 0, 0.3, 3.0, or 30 mg/kg/d INT131 Besylate by nasogastric gavage once daily for 26 weeks. Groups included 4 animals/sex/dose, with 2 additional animals/sex for 0 and 30 mg/kg/day groups for a 4 week recovery period.

Sprague-Dawley rats were treated with 0, 30, 100, or 300 mg/kg/d INT131 Besylate by gavage once daily for 26 weeks. Groups included 15 animals/sex/dose, with 5 additional animals/sex/ group which completed a 4 week recovery period after the dosing period.

Measurements for both studies included TK, body weight, clinical observations, hematology, clinical chemistry, urinalysis, organ weight, gross necropsy and microscopic observations. The monkey study also included ECG, ophthalmology exam, echocardiograms, and careful observation for edema.

Nonclinical Safety Study Exposure Relative to Clinical Exposure for INT131

Rat

Preclinical study dose (mg/kg/day)	male: 0.5	male: 1*	male: 2	male: 3	female: 0.5	female: 1*	female: 2	female: 3
30	397	198	99	66	2,066	1,033	508	344
100	907	454	227	151	4,546	2,273	1,118	758
300	1,829	914	457	305	6,216	3,108	1,529	1,036

(Column heads: Clinical study dose.)

Monkey

Preclinical study dose (mg/kg/day)	Clinical study dose: 0.5 mg	1 mg*	2 mg	3 mg
0.3	5	2	1	1
3	87	44	22	15
30	412	206	103	69

**1 mg dose met primary efficacy endpoint in Phase 2a study*

RESULTS

INT131 was well tolerated with no clinical observations of edema, signs of plasma volume expansion, significant cardiac hypertrophy, significant weight gain, hepatic toxicity, or fatty replacement of marrow in either rats or monkeys at any dose. The NOAEL was identified in monkey as the high dose (30 mg/kg/day), which translated to exposures of equal or greater than 70 fold the anticipated clinical exposure in the Phase 2b. Greater exposures were attained in the rat, with absence of typical PPARγ full agonist toxicities at 2-10 fold (males/ females) higher levels than in the high dose monkey group.

Figure 4: Body weight in rats and monkeys treated daily with high dose INT131

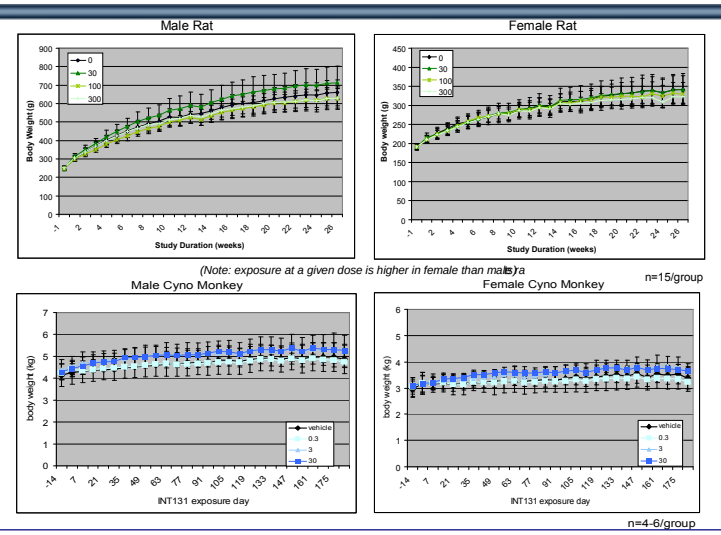
Male Rat
Female Rat
Body Weight (g)
Study Duration (weeks)
(Note: exposure at a given dose is higher in female than males)
n=15/group
Male Cyno Monkey
Female Cyno Monkey
body weight (kg)
INT131 exposure day
vehicle
0.3
3
30
n=4-6/group

Rat Study Findings

- Clinical observations
 - Unkempt appearance, high dose♀
 - No other notable observations
- Body weight
 - No significant differences
- Food consumption
 - No significant differences
- Clinical chemistry
 - No significant differences
- Hematology
 - No significant differences
- Coagulation parameters
 - No significant differences
- Urinalysis
 - No significant differences
- Gross necropsy
 - No significant differences
- Death of 3 high dose♀ (day 11, 49, 89); no findings, cause unknown

Cynomolgus Monkey Study Findings

- Echocardiogram
 - No significant differences
- ECG
 - No significant differences
- Food consumption
 - No significant differences
- Clinical chemistry
 - No significant differences
- Hematology
 - No significant differences
- Coagulation parameters
 - No significant differences
- Urinalysis
 - No significant differences
- Ophthalmology
 - No significant differences
- Clinical observations
 - No significant differences
 - Careful observation for edema
- Body weight
 - No significant differences
 - Trend to increase at mid and high dose early in study
 - Trend to loss at low dose early in study
- Gross necropsy
 - No significant differences
- Organ weight
 - No test article related changes
- Histology
 - No notable or adverse observations

Figure 5: Hematocrit in rats and monkeys treated daily with high dose INT131

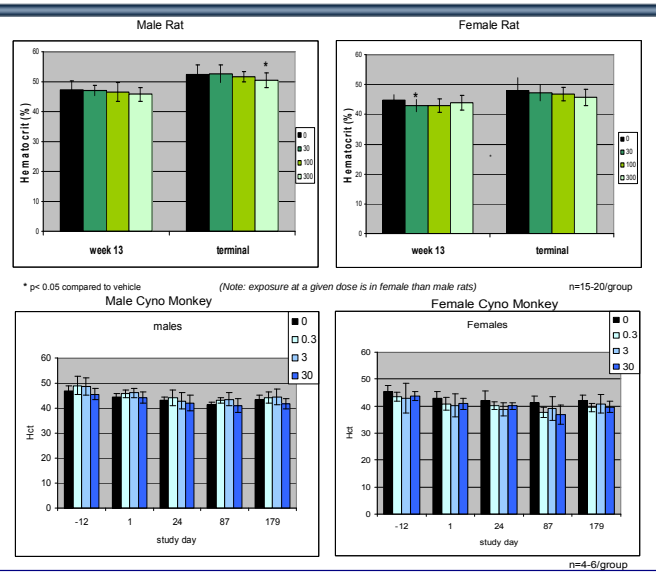
Male Rat
Female Rat
Hematocrit (%)
week 13
terminal
* p< 0.05 compared to vehicle
(Note: exposure at a given dose is in female than male rats)
n=15-20/group
Male Cyno Monkey
Female Cyno Monkey
males
Females
Hct
study day
n=4-6/group

Figure 6: Organ weight in rats and monkeys treated daily with high dose INT131

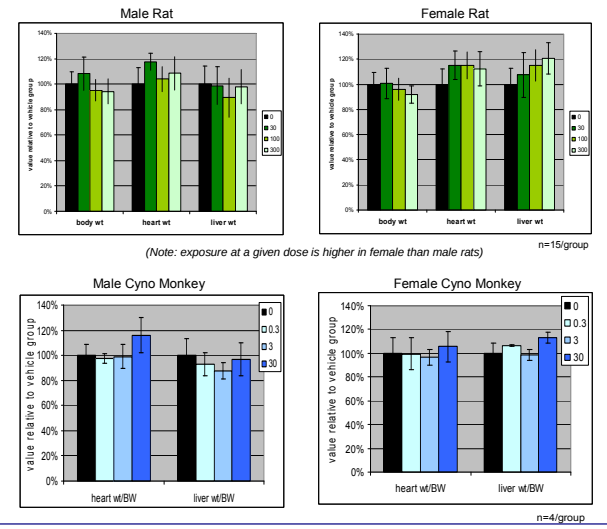
Male Rat
Female Rat
body wt
heart wt
liver wt
(Note: exposure at a given dose is higher in female than male rats)
n=15/group
Male Cyno Monkey
Female Cyno Monkey
value relative to vehicle group
heart wt/BW
liver wt/BW
n=4/group

Body weight and adiposity: (Figure 4) There were no significant differences treated rats or monkeys treated with high doses of INT131, though non-significant trends to increase were observed at the highest doses. There were no observations of increased subcutaneous adiposity, or increased white adipose tissue, or fatty infiltration of organs. A mild increase in intrascapular brown adipose tissue was observed in rat.

Fluid retention: (Figure 5) There were no observations of edema nor other evidence of fluid retention in rats or monkeys treated with high doses of INT131, consistent with lack of increased body weight. Modest but statistically significant reductions in hematocrit were seen in high dose male rats at 26 weeks and low dose female rats at 13 weeks, though the overall hematological profile in rats or monkeys did not support plasma volume expansion

Organ weight: (Figure 6) A mild, reversible increase in liver weight was observed in female rats was not considered adverse due to lack of histological or clinical chemistry evidence of pathology. No changes were found in male rats or in monkeys. A mild increase in heart weight in female rats was not deemed adverse due to small magnitude, lack of histological findings, and lack dose response. A mild increase in heart weight in high dose male monkeys was considered incidental due to lack of histological, echocardiogram, clinical chemistry findings, and variance in body size from vehicle group. There were no other changes in organ weight in either species.

No Adverse Effect Level (NOAEL)

- ***Rat*: 100 mg/kg/day, equivalent exposure to 420x(♂) and 2,100x(♀) exposure at efficacious dose in Phase 2a**
- ***Monkey*: 30 mg/kg/day, equivalent exposure to 210x exposure (♂and ♀) at efficacious dose in Phase 2a**

DISCUSSION

Six month nonclinical safety studies of PPARγ ligands have been shown to be highly predictive of clinical experience. Thus, six month rat and monkey studies were conducted using high doses of selective PPARγ modulator (SPPARM) INT131 to determine whether the separation of efficacy and side effect dose response curves predicted from the design and the in vitro and pharmacology selection of this moiety would be confirmed in the toxicology setting. Indeed, typical TZD effects observed in both toxicology studies and clinical experience, such as weight gain, fluid retention, and cardiovascular pathology were not seen with INT131 even at the highest doses tested, which provided exposures 2-3 orders of magnitude over clinical exposures. Clear differentiation from TZD full agonists was demonstrated: while the NOAEL in rats for Actos is 4.6x (♂) and for Avandia is 6.4x (♂) the exposure at approved clinical doses, for INT131 it is 420x (♂) and 2,100x (♀) the exposure at the similarly efficacious dose in a Phase 2a study. While the NOAEL in monkeys for Actos is 4.9x (not investigated with Avandia) the exposure at approved clinical doses, for INT131 is 210x the exposure at the similarly efficacious dose in a Phase 2a study.

CONCLUSIONS

INT131 demonstrated no significant fluid retention, weight gain, cardiac hypertrophy, or hepatic toxicity in cynomolgus monkeys or in rats treated for six months at dose exposure multiples of equal or greater than 70 times the anticipated clinical exposure in the highest dose in an ongoing Phase 2b study in subjects with T2DM. These results support in-vitro studies, rodent studies, and the clinical data demonstrating that INT131 is a SPPARM with unique PPARγ activating properties which minimize the commonly recognized side effects seen with full PPARγ agonists. These data support further study of INT131 as a potent insulin sensitizer for the treatment of T2DM without the significant side effects of the TZDs.

REFERENCES

[1] Li Y, Wang Z, Motani A, McGee L, Learned M, Timmermans, P, Chen JL. **T0903131 (T131): a Selective Modulator of PPARγ,** Abstract 659 ADA 2004

[2] F Dunn, L Higgins, A DePaoli, **INT131: A Selective PPARγ Modulator (SPPARM) for Type 2 Diabetes Mellitus** Abstract 499-P ADA 2008

[3] Team leader memo to file regarding preclinical pharmacology/toxicology issues for NDA 20-073 (PIOGLITAZONE, ACTOS®)

[4] CDER pharmacological review of application 021071 (ROSIGLITAZONE, AVANDIA ®)

[5] El Hage J. Peroxosome Proliferator-Activated Preceptor Agonists: Preclinical and Clinical Cardiac Safety Considerations. 2005. http://www.fda.gov/ohrms/dockets/ac/05/slides/2005-4169S2_02_02-FDA-ElHage.ppt. Accessed 2008.